Poster 242

A phase 1 multicenter, dose escalation study of CBT-501, a novel anti-PD-1 inhibitor in subjects with select advanced or relapsed/recurrent solid tumors

Purvi Patel[1], Mamatha Reddy[1], Melissa Lopez[1], Neil Sankar[1], Sarath Kanekal[1], Mike Li[1], Sanjeev Redkar[1] and Gavin Choy[1]

[1]CBT Pharmaceuticals, Inc., Pleasanton, CA, USA.

BACKGROUND

- Checkpoint blockades, such as the Programmed Death-1 (PD-1) / Programmed Death Ligand 1 (PD-L1) pathway regulate the progression of productive immune responses.
- Blocking the PD-1 / PD-L1 pathway can increase the function of effector CD8+ T cells and enhance the anti-tumor response.
- Monoclonal antibodies (mAbs) to block the PD pathway is a clinically validated approach for cancer immunotherapy.

CBT-501

- CBT-501, genolimzumab (GB226), is a novel humanized IgG4 mAb targeting the PD-1 membrane receptor on T cells with a highly specific binding to PD-1 of human (Kd=505 pM) and cynomolgus monkeys (Kd=7.2 nM)[1].
- CBT-501 occupancy in activated CD4+ and CD8+ T-cells increases rapidly after a single dose **(Figure 1)** and upon multi-dosing, receptor occupancy rises to nearly 100% **(Figure 2)**.[1] Six weeks after the last dose, occupancy on CD4+ and CD8+ cells does not significantly decline.
- CBT-501 has demonstrated anti-tumor activity in the in vivo animal model and no abnormal drug-related toxicity has been observed in the GLP toxicology studies.[1] **(Figure 3)**
- CBT-501 demonstrated dose dependency with a half-life of 5 to 10 days and exhibited linear pharmacokinetic features in the in-vivo monkeys[1]. **(Figure 4)**
- Data from all pre-clinical pharmacodynamics and toxicology studies of CBT-501 indicate pharmacological activity at effective doses with a wide margin of safety.
- Based on these findings, CBT has initiated a Phase 1 study in Australia with CBT-501 for subjects with select advanced or relapsed/recurrent solid tumors. Genor, CBT's China partner, has also initiated a Phase 1 trial in China.

PRE-CLINICAL RATIONALE

Figure 1: Single Dose Receptor Occupancy on Activated CD4+ and CD8+ T Cells in Cynomolgus Monkeys

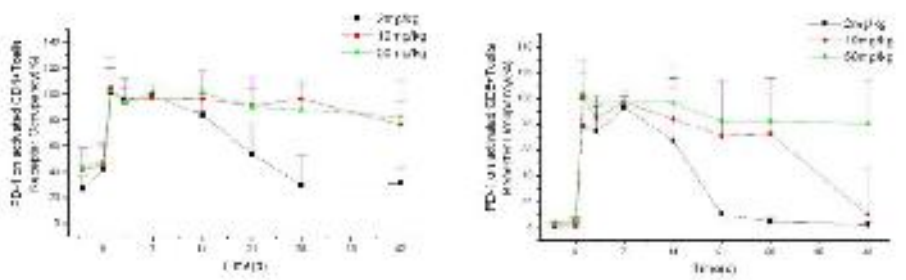

Figure 2: Multi-Dose Receptor Occupancy on Activated CD4+ and CD8+ T Cells in Cynomolgus Monkeys

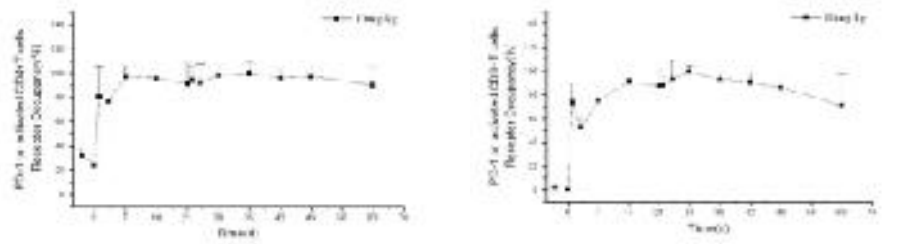

Figure 3: Humanized Genetically Engineered (huGEMM) MC38 Mouse Colon Adenocarcinoma Demonstrates Anti-tumor Activity Comparable to Nivolumab

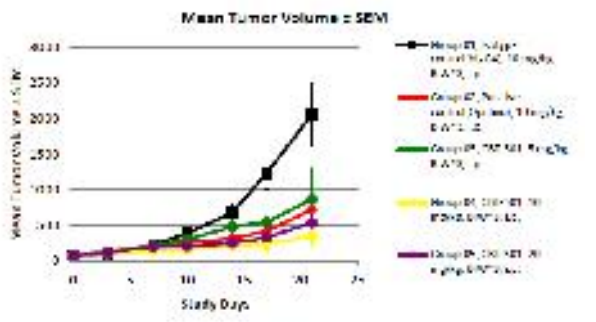
Mean Tumor Volume ± SEM
Study Days

Figure 4: Pharmacokinetics after Single Dose and Repeat Doses in Cynomolgus Monkeys

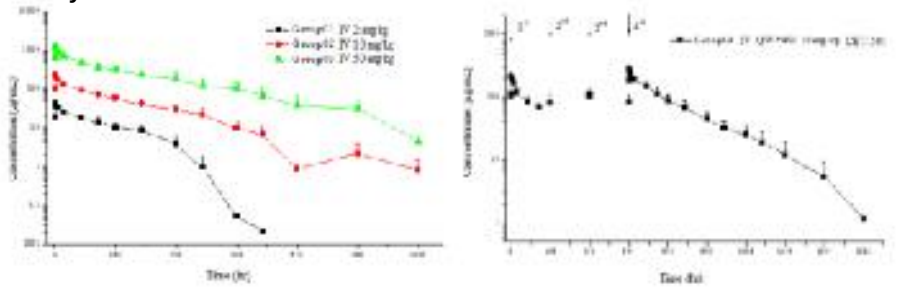

PHASE 1 STUDY METHODS

- CBT-501-01 is a Phase 1, multicenter, dose escalation study of CBT-501 in subjects in select advanced or relapsed/recurrent solid tumors, including cHL, currently in progress in Australia (NCT03053466).
- The primary study objective is to identify the overall safety and tolerability, including any dose limiting toxicities (DLT), and determine the recommended Phase 2 dose (RP2D).
- Secondary study objectives include assessing efficacy by overall response rate (ORR), best overall response rate (BOR) per RECIST v1.1 and irRECIST, time to response, duration of response (DOR), disease control rate (DCR) by RECIST v1.1 and irRECIST, progression free survival (PFS), and determining the pharmacokinetic (PK) parameters.
- Exploratory objectives involve the assessment of PD-1 and PD-L1 expression, receptor occupancy and the host immune response in peripheral blood or formalin-fixed paraffin-embedded (FFPE) samples.
- This is a 3-part study with a dose-escalation segment, cohort extension, and dose and disease expansion cohorts. Up to 82 subjects will be enrolled.

CLINICAL STUDY DESIGN

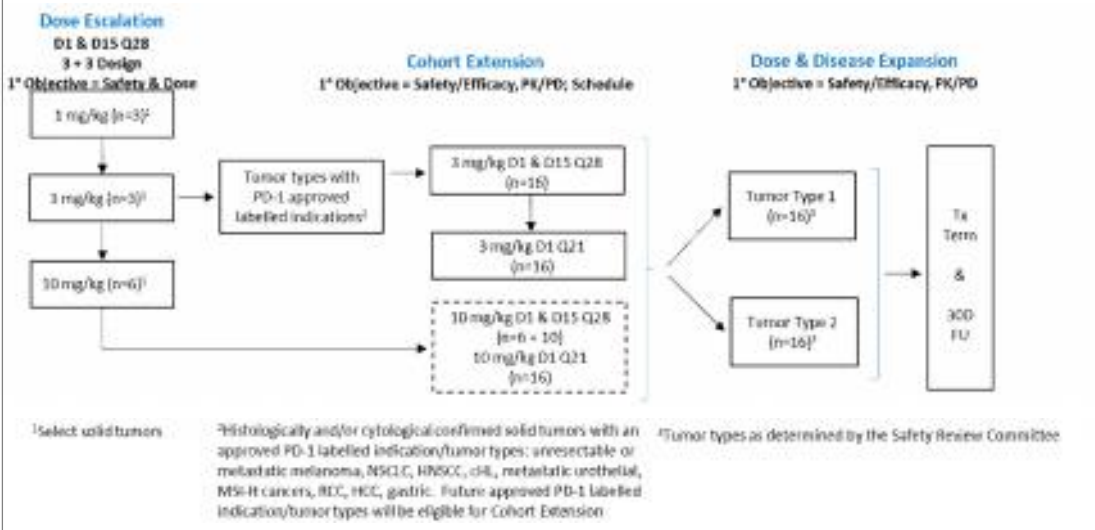
Dose Escalation
D1 & D15 Q28
3 + 3 Design
1° Objective = Safety & Dose
1 mg/kg (n=3)¹
3 mg/kg (n=3)¹
10 mg/kg (n=6)¹
Cohort Extension
1° Objective = Safety/Efficacy, PK/PD; Schedule
Tumor types with PD-1 approved labelled indications²
3 mg/kg D1 & D15 Q28 (n=16)
3 mg/kg D1 Q21 (n=16)
10 mg/kg D1 & D15 Q28 (n=6 + 10)
10 mg/kg D1 Q21 (n=16)
Dose & Disease Expansion
1° Objective = Safety/Efficacy, PK/PD
Tumor Type 1 (n=16)³
Tumor Type 2 (n=16)³
Tx Term & 30D FU
¹Select solid tumors
²Histologically and/or cytological confirmed solid tumors with an approved PD-1 labelled indication/tumor types: unresectable or metastatic melanoma, NSCLC, HNSCC, cHL, metastatic urothelial, MSI-H cancers, RCC, HCC, gastric. Future approved PD-1 labelled indication/tumor types will be eligible for Cohort Extension
³Tumor types as determined by the Safety Review Committee

ELIGIBILITY CRITERIA

MAJOR INCLUSION	MAJOR EXCLUSION
▪ ≥ 18 years of age	• Hypersensitivity to mAbs
▪ Confirmation of select solid tumor	• Active malignancy within 2 years
▪ ECOG 0 - 1	• Prior therapy with antibody targeting T cell co-stimulation
▪ Life expectancy > 3 months	• Known autoimmune disease
▪ Measurable disease per RECIST v1.1	• Known significant mental illness

PARTICIPATING STUDY CENTERS

- **Australia, New South Wales**
 Chris O'Brien Lifehouse
 Principal Investigator: Catriona McNeil, MBBS
- **Australia, Victoria**
 Cabrini Education and Research Precinct
 Principal Investigator: Gary Richardson, MBBS
 Nucleus Network
 Principal Investigator: Mark Voskoboynik, MBBS
- **Australia, Western Australia**
 Linear Clinical Research
 Principal Investigator: Michael Millward, MBBS

REFERENCE

1. Qing Z, Nian W, Sun Z et al. CBT-501, a novel humanized anti-PD-1 antibody shows differentiated activity in a humanized genetically engineered mouse model of colon carcinoma. ASCO-SITC, 2017, Abstract 108.

FURTHER INFORMATION

Please visit CBT Pharmaceutical's website at www.cbtpharma.com for a PDF version of the poster presentation.

Society for Immunotherapy of Cancer (SITC), November 10-12, 2017, Gaylord National Hotel & Convention Center in National Harbor, MD